A Phase 1 Trial of IO-202, An Antagonist Antibody Targeting Myeloid Checkpoint LILRB4 (ILT3), as Monotherapy and in Combination with Pembrolizumab in Adult Patients with Advanced Relapsed or Refractory Solid Tumors

IMMUNE-ONC therapeutics

SITC Poster #747

Aung Naing[1], MD, John Powderly[2], MD, Meredith S. Pelster[3], MD, MSCI, Alexander I Spira[4], MD, PhD, Reva E. Schneider[5], MD, Paul Woodard[6], MD, Luke Chung[6], MD, Elizabeth Wieland[6], MS, Sydney Ray[6], MS, Kyu Hong[6], MS, Tao Huang[6], PhD, X. Charlene Liao[6], PhD, Hong Xiang[6], PhD, Wen Hong Lin[6], MD, MS and Heinz-Josef Lenz[7], MD

[1]Anderson Cancer Center, Houston, TX; [2]Carolina BioOncology Institute, Huntersville, NC; [3]Sarah Cannon Research Institute at Tennessee Oncology, Nashville, TN; [4]Virginia Cancer Specialists, Fairfax, VA, [5]Mary Crowley Cancer Institute, Dallas, TX, [6]Immune-Onc Therapeutics, Inc., Palo Alto, CA, [7]USC Norris Comprehensive Cancer Center, Los Angeles, CA

1 Background

Most patients with advanced solid tumors relapse after T-cell checkpoint blockade, despite of immunotherapy becoming the mainstream with the approvals of T-cell checkpoint inhibitors. Therefore, there is still an unmet medical need. Myeloid checkpoint inhibition is a new approach to cancer immunotherapy.

Leukocyte immunoglobulin-like receptor subfamily B member 4 (LILRB4) is a myeloid checkpoint with its expression restricted to monocytes or monocyte-derived cells[1,2,3] and in normal antigen presenting cells. LILRB4 functions as a negative regulator of immunity through interaction with its ligands, apolipoprotein E and fibronectin [1,2,4,5,6]. Blockade of LILRB4 has the potential to enhance anti-tumor T cell activities.

IO-202 is a IgG1 monoclonal antibody that binds to LILRB4 to block ligand interactions and inhibits the function of LILRB4. In vitro, IO-202 treatment of immune cells increases pro inflammatory responses and enhances antigen presenting cell phenotypes. IO-202 has been studied at a first-in-human, phase 1 study in acute myeloid leukemia and chronic myelomonocytic leukemia patients (IO-202-CL-001) up to 30 mg/kg IV Q2W with no observed dose limiting toxicity (DLT), which provided sufficient data supporting the starting dose of 250 mg in solid tumors (IO-202-CL-002 trial).

2 Targeting Myeloid Cells to Overcome Limitations of Current Treatment Options

Myeloid cells modulate key cancer-associated activities, including ***immune evasion***, and affect virtually all types of cancer therapy

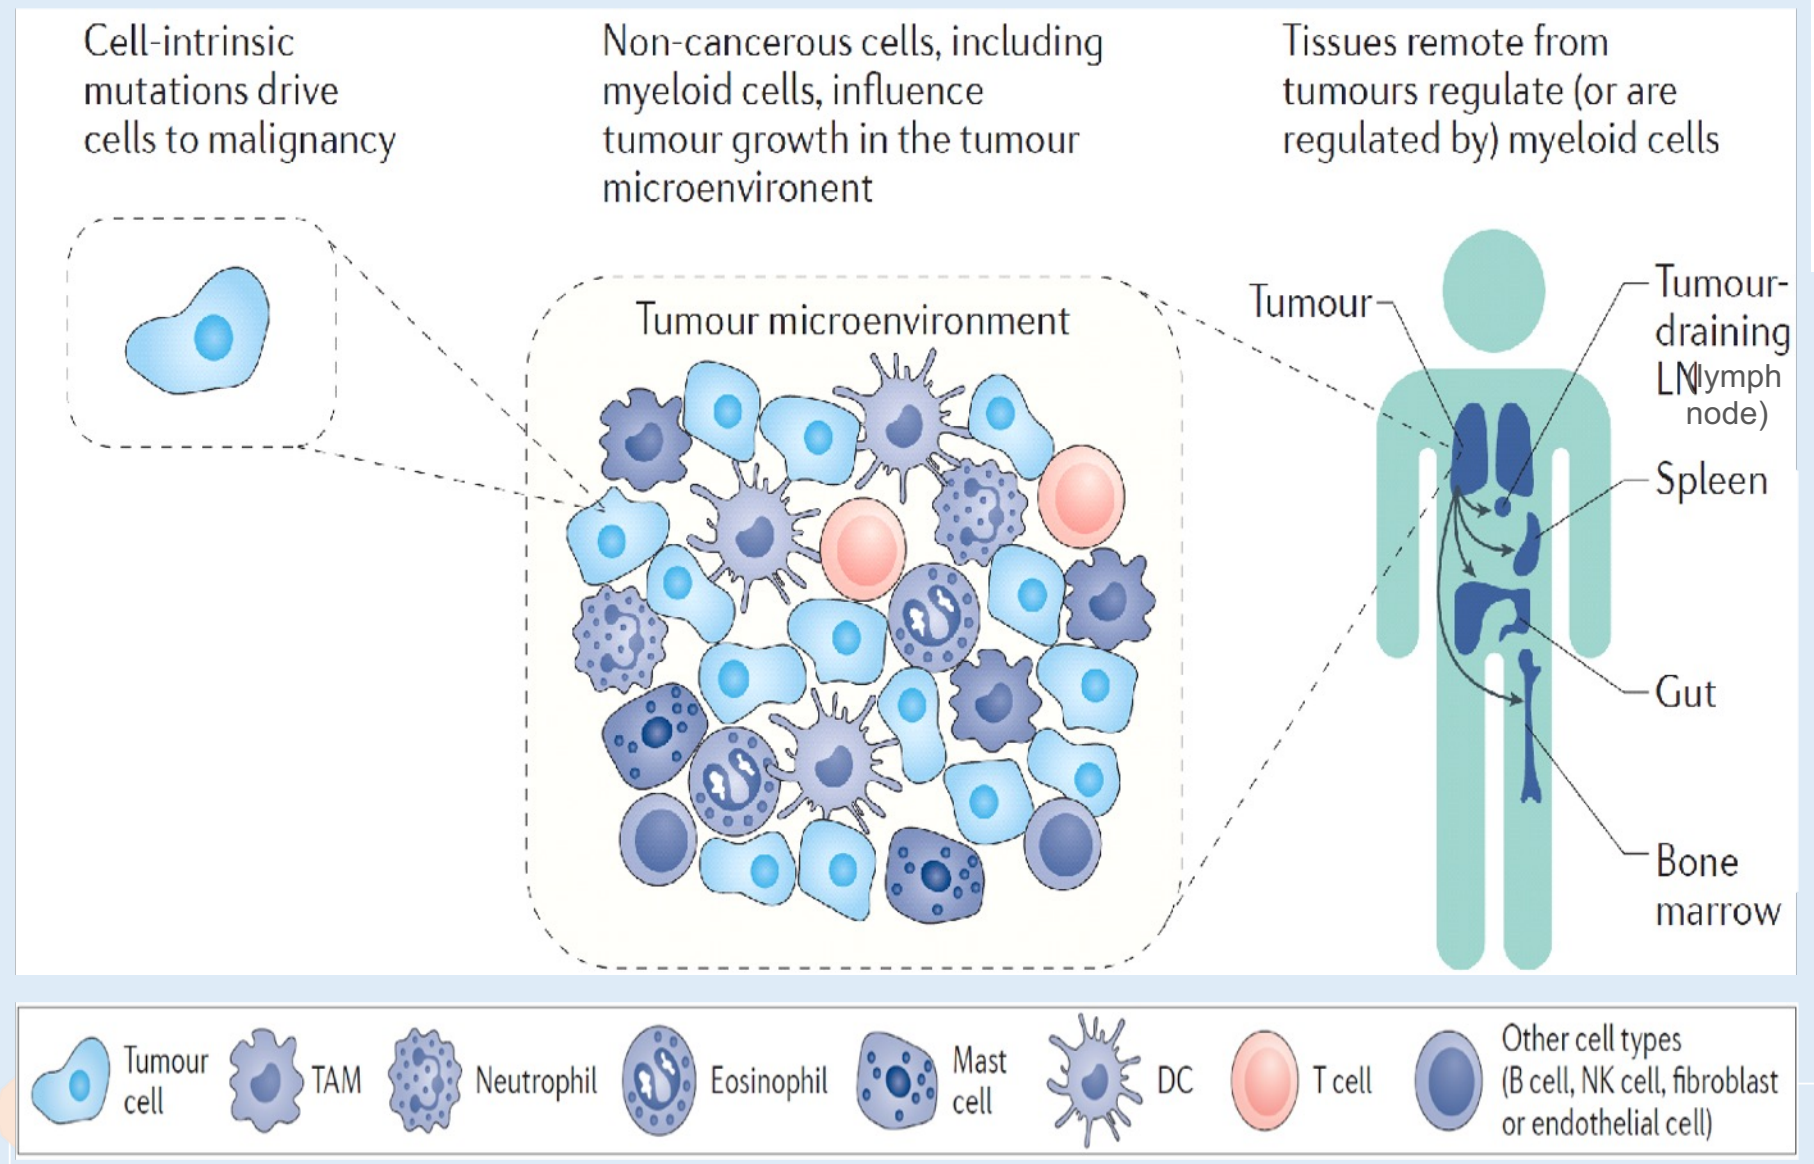
Cell-intrinsic mutations drive cells to malignancy
Non-cancerous cells, including myeloid cells, influence tumour growth in the tumour microenvironent
Tissues remote from tumours regulate (or are regulated by) myeloid cells
Tumour microenvironment
Tumour
Tumour-draining LN (lymph node)
Spleen
Gut
Bone marrow
Tumour cell
TAM
Neutrophil
Eosinophil
Mast cell
DC
T cell
Other cell types (B cell, NK cell, fibroblast or endothelial cell)

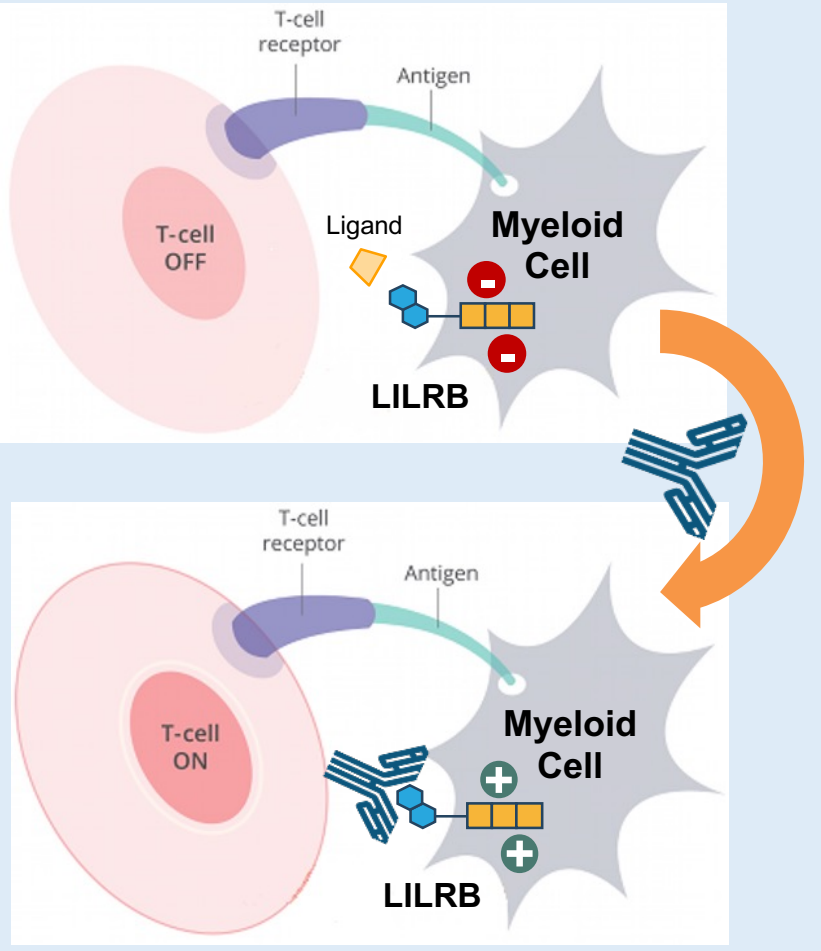
T-cell receptor
Antigen
T-cell OFF
Ligand
Myeloid Cell
LILRB
T-cell ON
Myeloid Cell
LILRB

Blocking Myeloid Checkpoints Will:
- Increase antigen presentation
- Reprogram myeloid phenotypes
- Release pro-inflammatory cytokines
- Attract and activate effector cells (T, NK, etc.)

Adapted from Nat Rev Cancer. 16:447-62 (2016)

LILRB4 Activation in Myeloid Cells Promotes Immune Inhibitory Signals

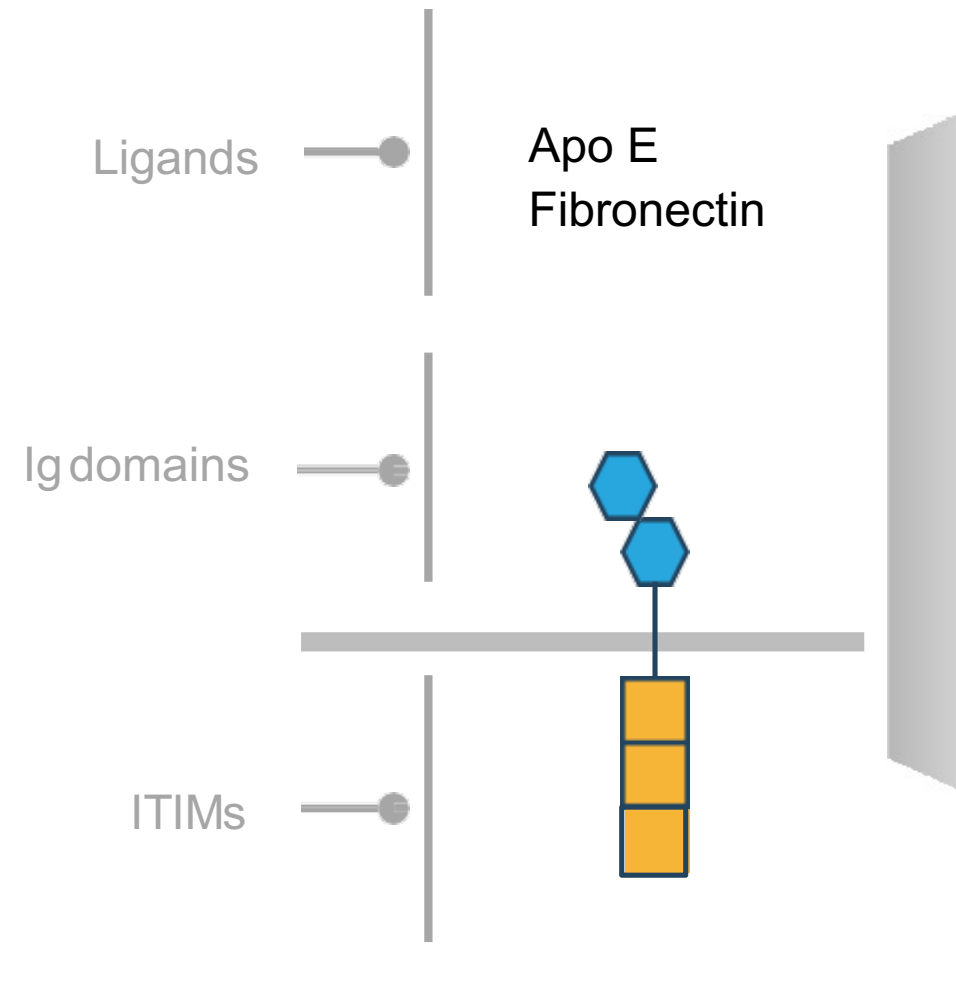
Ligands
Apo E
Fibronectin
Ig domains
ITIMs

Cell Intrinsic: LILRB4 inhibits antigen presenting cell activation

Cell Extrinsic: LILRB4 induces immune tolerance via T-suppressor cells

LILRB4 does not exist in mouse

ITIM = immunoreceptor tyrosine-based inhibition motif

Cancer Immunol Res (2021)
Nature 562, 605-609 (2018)
J Immunol 176, 2790-2798 (2006)
Nat Immunol 3, 237-243 (2002)
J Exp Med 185, 1743-1751 (1997)

3 IO-202 Mechanism of Action

Proposed Mechanism of Action

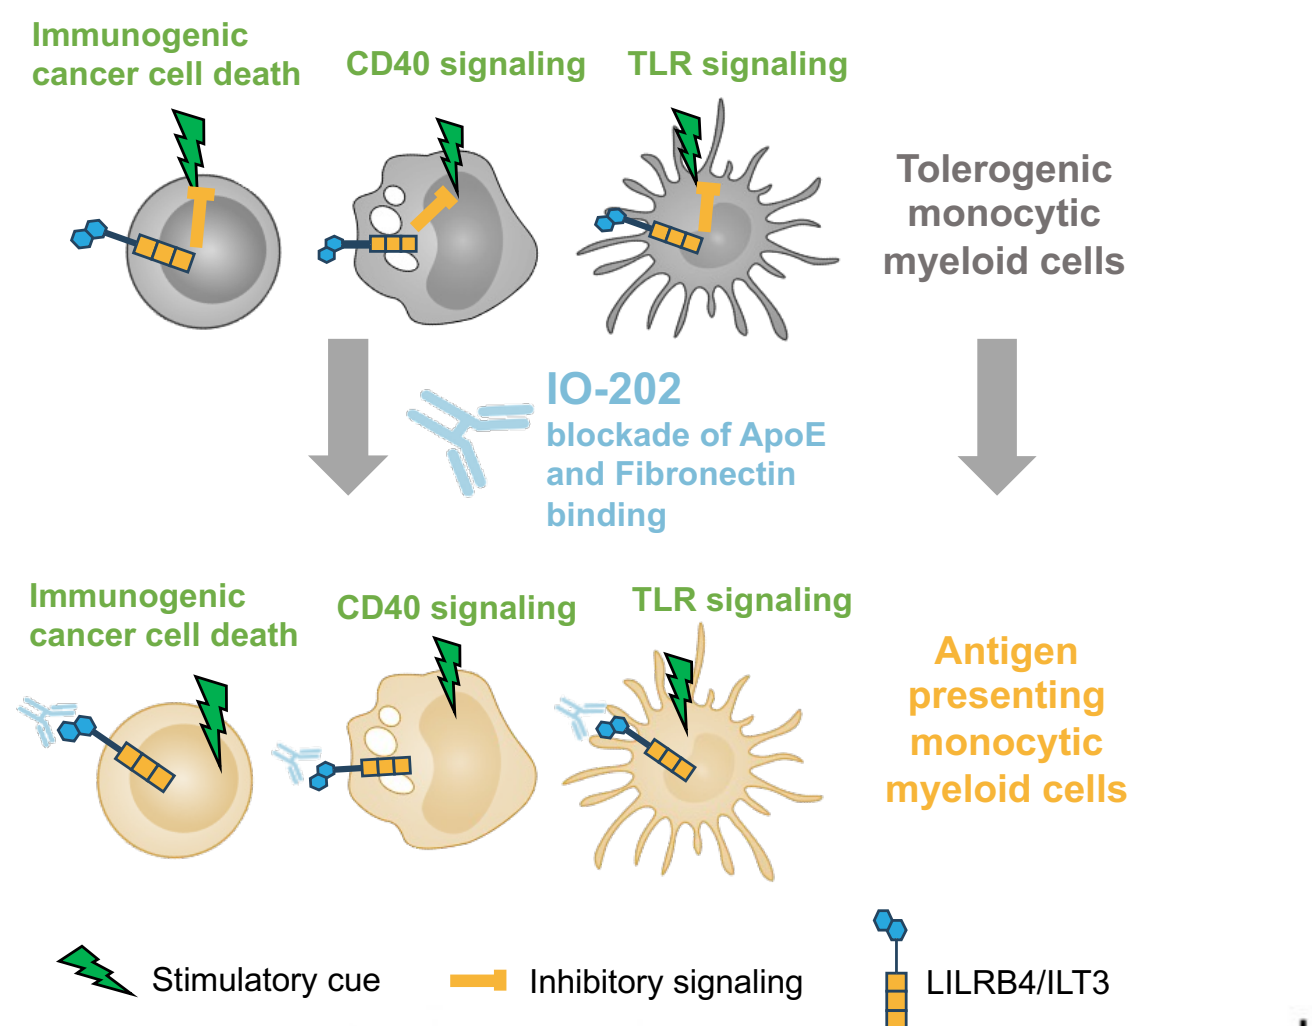
Immunogenic cancer cell death
CD40 signaling
TLR signaling
Tolerogenic monocytic myeloid cells
IO-202 blockade of ApoE and Fibronectin binding
Immunogenic cancer cell death
CD40 signaling
TLR signaling
Antigen presenting monocytic myeloid cells
Stimulatory cue
Inhibitory signaling
LILRB4/ILT3

LILRB4 Reporter cell

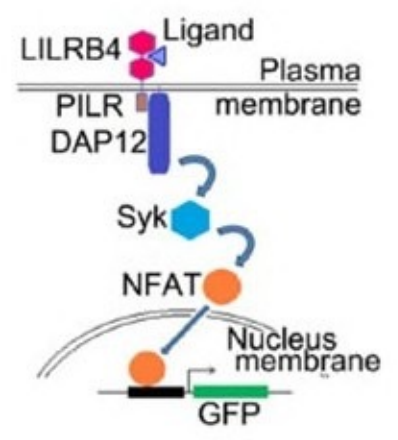
LILRB4
Ligand
Plasma membrane
PILR
DAP12
Syk
NFAT
Nucleus membrane
GFP

In Vivo Activity in Radiation Therapy (RT)-Resistant Model

Lewis lung carcinoma in syngeneic *LILRB4* transgenic mice

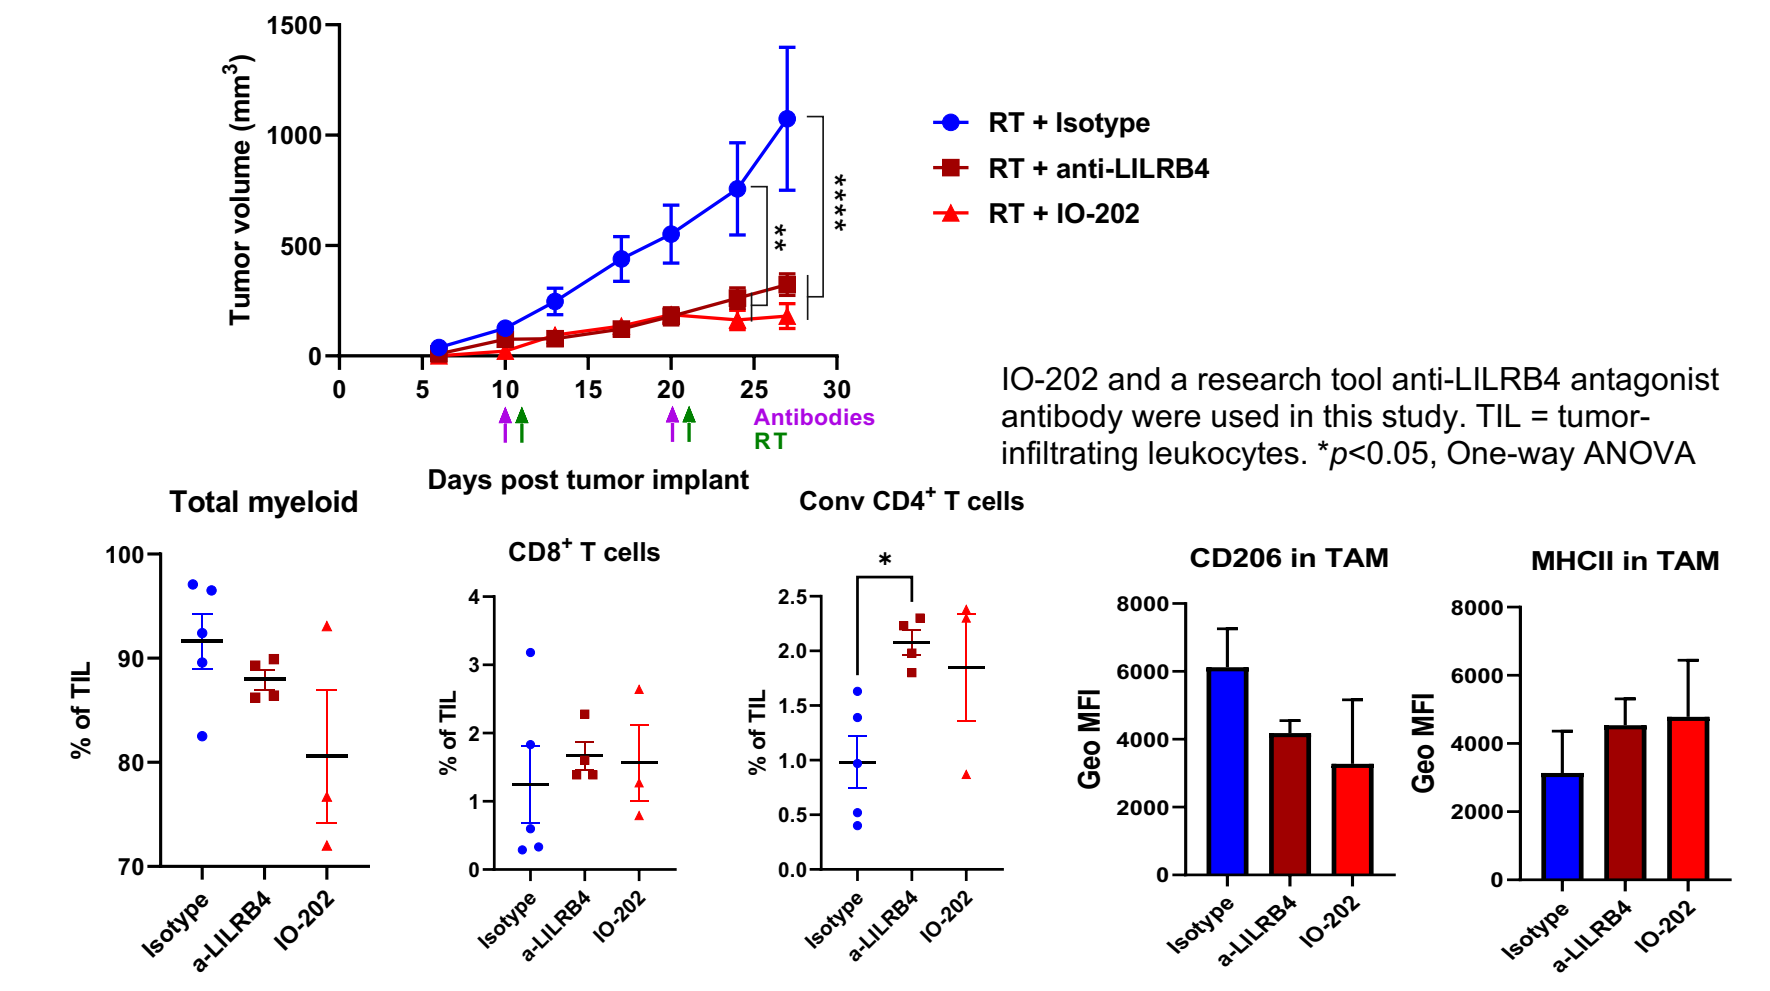
Tumor volume (mm3)
Days post tumor implant
Antibodies
RT
RT + Isotype
RT + anti-LILRB4
RT + IO-202
Total myeloid
CD8+ T cells
Conv CD4+ T cells
CD206 in TAM
MHCII in TAM
% of TIL
Geo MFI
Isotype
a-LILRB4
IO-202

IO-202 and a research tool anti-LILRB4 antagonist antibody were used in this study. TIL = tumor-infiltrating leukocytes. *p<0.05, One-way ANOVA

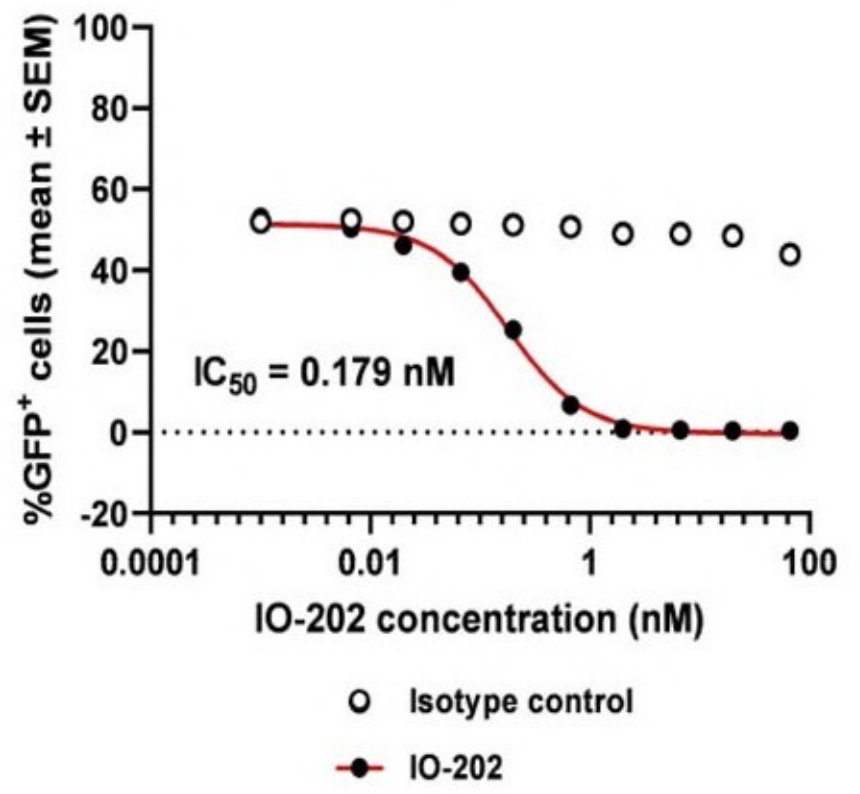
Ligand: APOE
%GFP+ cells (mean ± SEM)
IC50 = 0.179 nM
IO-202 concentration (nM)
Isotype control
IO-202

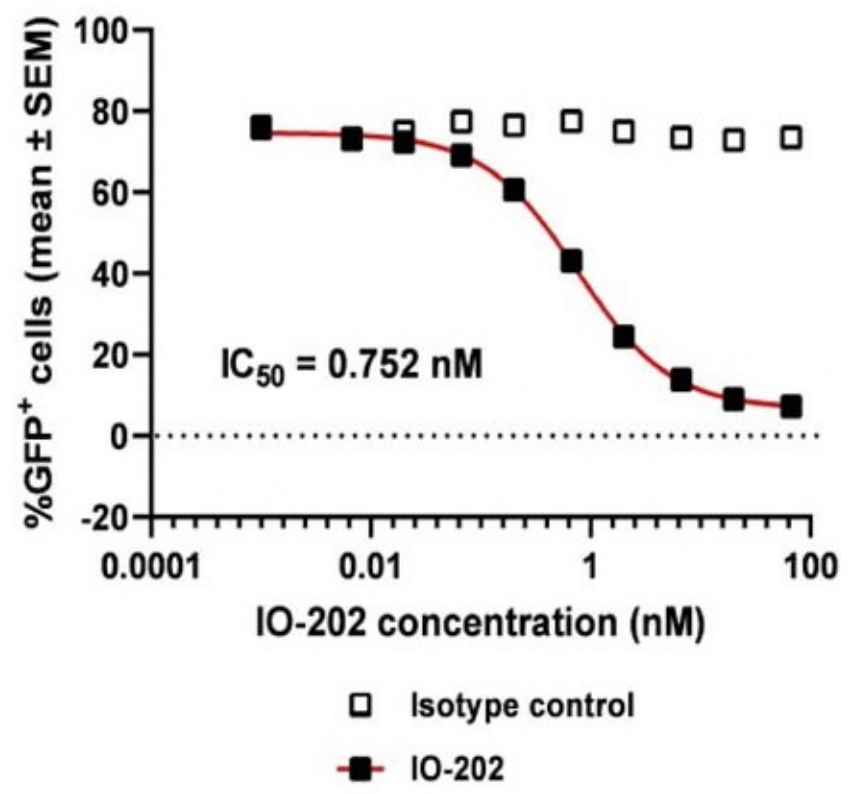
Ligand: FN
%GFP+ cells (mean ± SEM)
IC50 = 0.752 nM
IO-202 concentration (nM)
Isotype control
IO-202

4 Phase 1 Clinical Study Objectives

Primary
- To assess safety and tolerability at increasing dose levels of IO-202 in successive cohorts of participants with advanced relapsed or refractory solid tumors in order to estimate the maximum tolerated dose (MTD) or maximum administered dose (MAD) as monotherapy and in combination with pembrolizumab, and select the recommended Phase 2 dose (RP2D)

Secondary
- To characterize the PK of IO-202
- To evaluate the immunogenicity of IO-202 and its impact on other endpoints
- To evaluate preliminary anti-tumor activity of IO-202

Exploratory
- To characterize potential PD effects of IO-202 in the periphery and tumor microenvironment

5 Phase 1 Clinical Study Design

This clinical trial is a first-in-human, Phase 1, multicenter, open-label, dose-escalation, safety, pharmacokinetic (PK), and pharmacodynamic evaluation of intravenously administered IO-202, both as monotherapy and in combination with pembrolizumab, in adult patients with solid tumors that have failed standard of care therapies. It is estimated that up to 36 patients will be enrolled in the dose escalation portion of the study. IO-202 will be administered every 21 days, with a dose-limiting toxicity (DLT) evaluation period of 21 days. Patients will be enrolled into sequential cohorts and treated with increasing doses of IO-202 using mTPI (modified Toxicity Probability Interval) method. Combination therapy will be initiated after the first dose level in monotherapy is cleared. Safety, PK, and clinical activity will be evaluated on an ongoing basis.

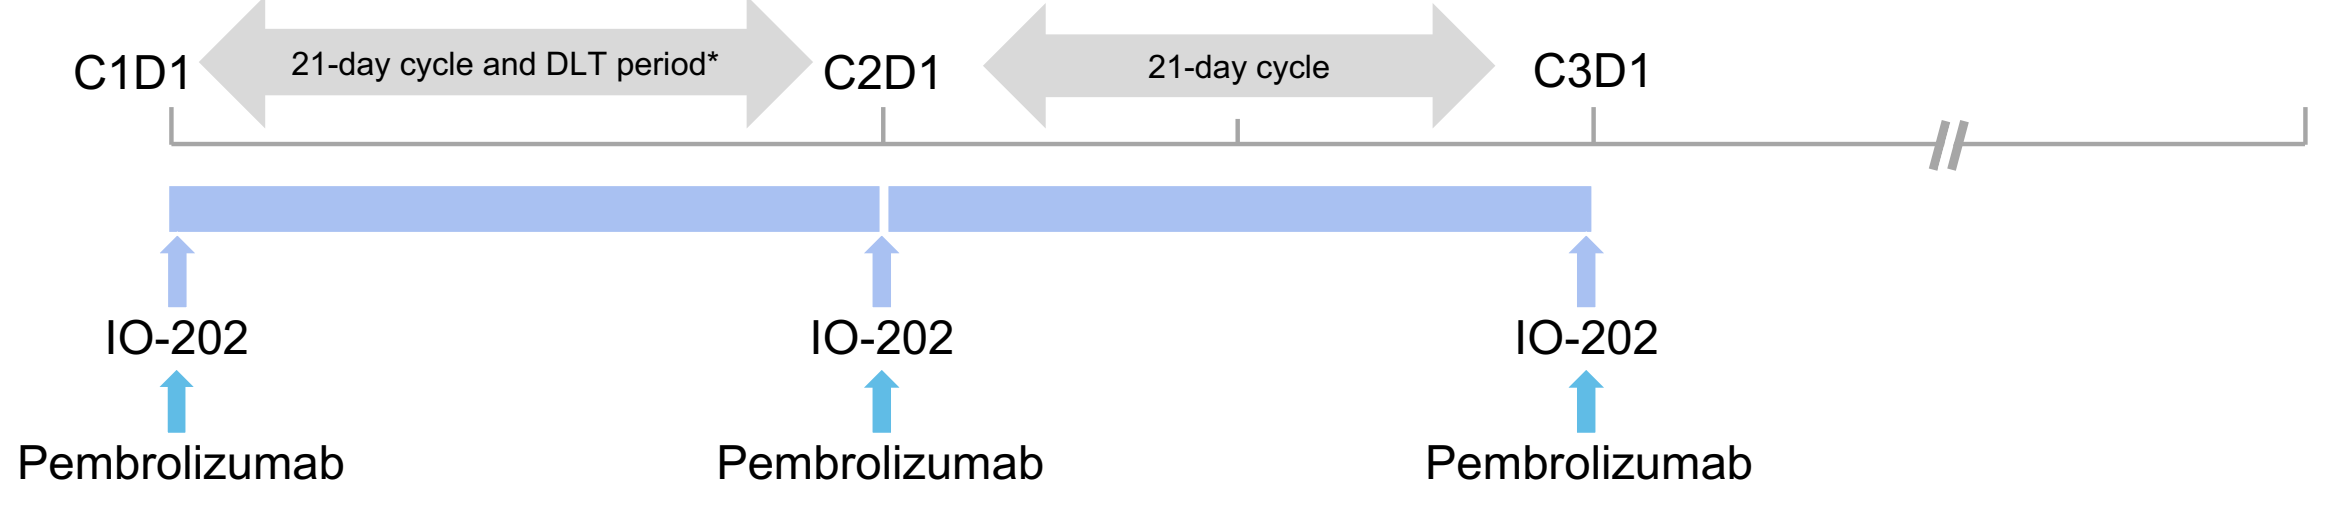
C1D1
21-day cycle and DLT period*
C2D1
21-day cycle
C3D1
IO-202
Pembrolizumab
IO-202
Pembrolizumab
IO-202
Pembrolizumab

*DLT (dose limiting toxicity) is assessed during 1st Cycle only

6 Dose Escalation and Dose Expansion Schema

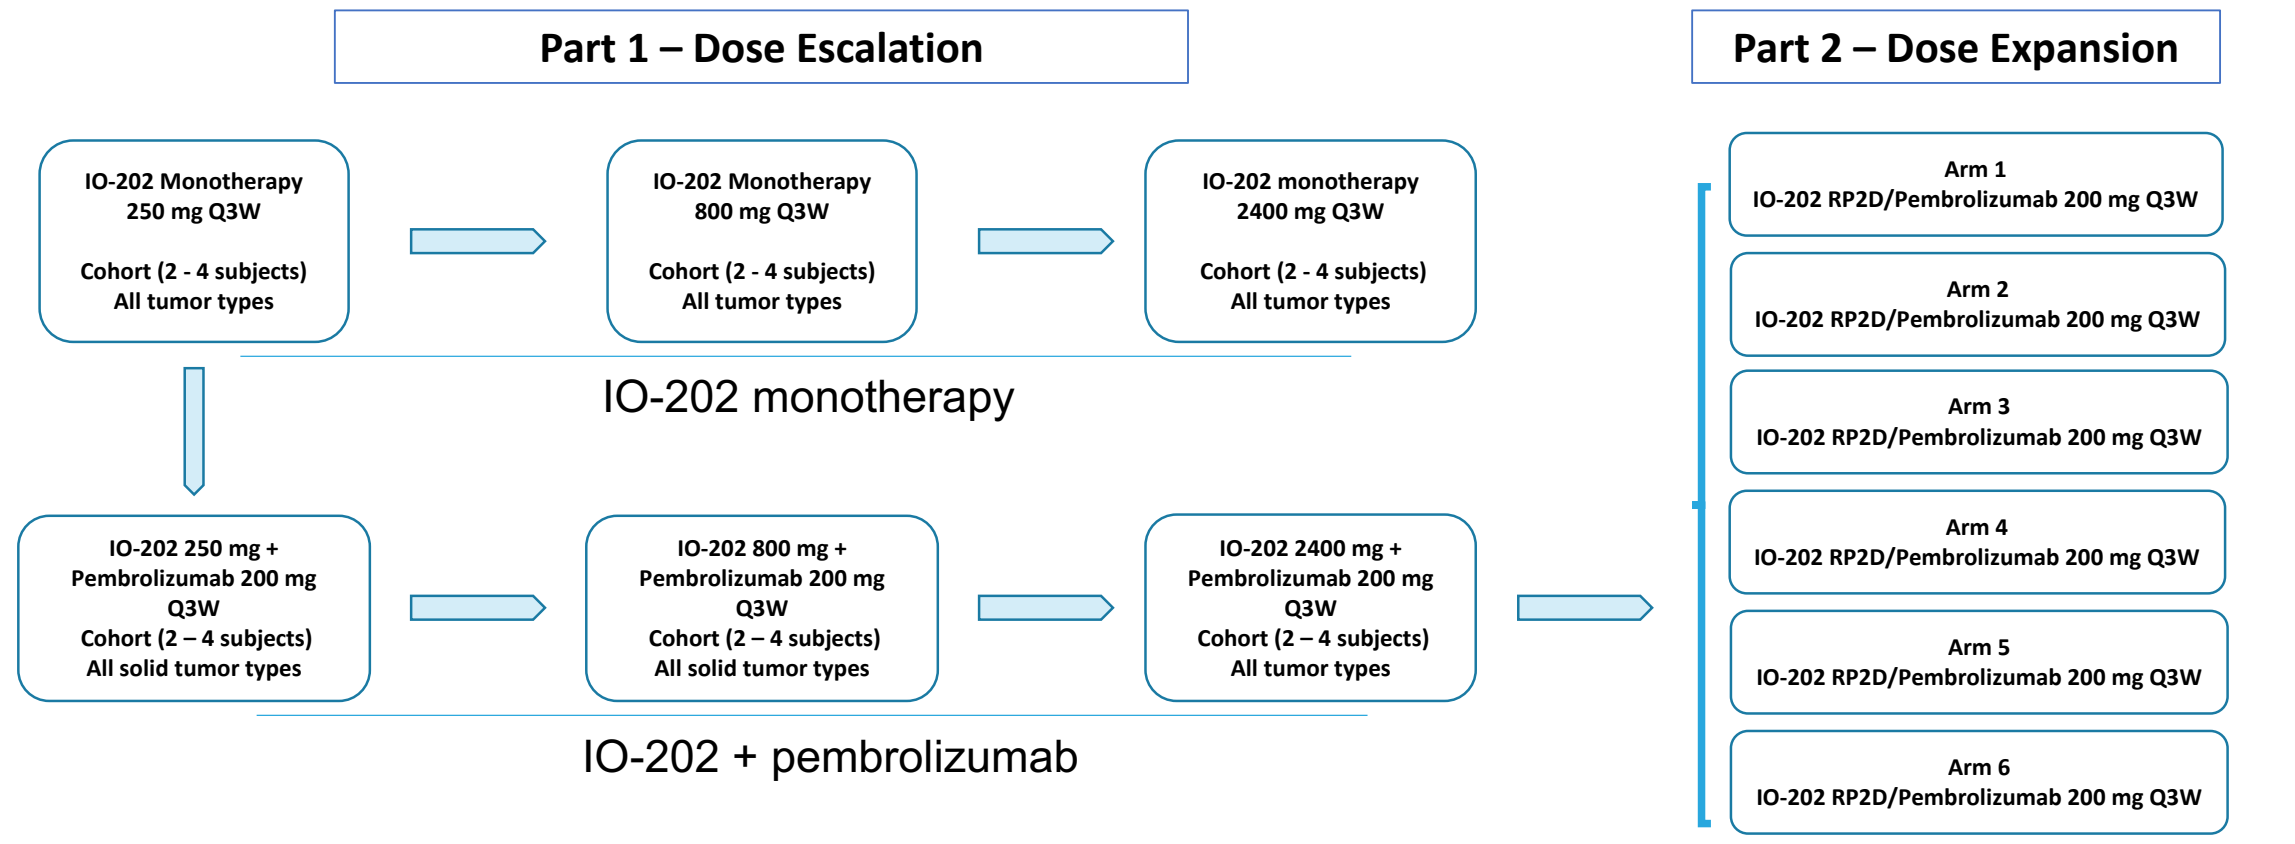
Part 1 – Dose Escalation
Part 2 – Dose Expansion
IO-202 Monotherapy 250 mg Q3W Cohort (2 - 4 subjects) All tumor types
IO-202 Monotherapy 800 mg Q3W Cohort (2 - 4 subjects) All tumor types
IO-202 monotherapy 2400 mg Q3W Cohort (2 - 4 subjects) All tumor types
IO-202 monotherapy
IO-202 250 mg + Pembrolizumab 200 mg Q3W Cohort (2 – 4 subjects) All solid tumor types
IO-202 800 mg + Pembrolizumab 200 mg Q3W Cohort (2 – 4 subjects) All solid tumor types
IO-202 2400 mg + Pembrolizumab 200 mg Q3W Cohort (2 – 4 subjects) All tumor types
IO-202 + pembrolizumab
Arm 1 IO-202 RP2D/Pembrolizumab 200 mg Q3W
Arm 2 IO-202 RP2D/Pembrolizumab 200 mg Q3W
Arm 3 IO-202 RP2D/Pembrolizumab 200 mg Q3W
Arm 4 IO-202 RP2D/Pembrolizumab 200 mg Q3W
Arm 5 IO-202 RP2D/Pembrolizumab 200 mg Q3W
Arm 6 IO-202 RP2D/Pembrolizumab 200 mg Q3W

7 Key Eligibility

- Age ≥18 years-old
- Have any histologically- or cytologically confirmed advanced/metastatic solid tumor (any type) by pathology report and received, been intolerant to, or been ineligible for standard systemic therapy known to confer clinical benefit. Patients with asymptomatic CNS disease allowed. (Dose Escalation)
- Has had stable disease for ≥6 months, a partial response or complete response on previous PD-1/PD-L1 inhibitor-containing therapy (Dose Escalation)
- Failed at least one available therapy for the disease under study (Dose Expansion)
- Has measurable disease by Response Evaluation in Solid Tumors version 1.1 (RECIST 1.1) as assessed by local site

8 Biomarker Plan

- IO-202 target engagement by determining LILRB4 receptor occupancy in peripheral blood myeloid cells
- Potential PD effects of IO-202 monotherapy and IO-202 + pembrolizumab in the TME and periphery:
 - Myeloid cell phenotyping
 - T cell profiling
 - Gene expression profiles of tumor tissues
- Correlations between baseline LILRB4 expression and tumor gene expression profile with safety, clinical activity, PK and PD of IO-202 monotherapy and IO-202+ pembrolizumab

9 Summary

- Myeloid checkpoint inhibition through blockade of the LILRB4 pathway has the potential to reactivate or enhance anti-tumor T cell immune responses.
- IO-202 is a fully human IgG1 anti-LILRB4 antibody displaying high affinity, specificity and potent antagonistic activity that promotes pro-inflammatory phenotype and differentiation of myeloid cells, enhances immune cell activation and alleviates myeloid cell suppressive activity
- This is a first-in-human study of IO-202 in solid tumor patients as monotherapy or in combination with pembrolizumab to evaluate safety/tolerability and determine the MTD/RP2D
- Key eligibility include patients ≥18 years with confirmed advanced/metastatic solid tumors of any type who have received or are ineligible for standard of care therapies

References:
1. Chang C, Ciubotariu R, Manavalan J, et al. Tolerization of dendritic cells by T(S) cells: the crucial role of inhibitory receptors ILT3 and ILT4. Nat Immunol. 2002;3:237-243.
2. Deng M, Gui X, Kim J. et al. LILRB4 signalling in leukaemia cells mediates T cell suppression and tumour infiltration. Nature. 2018; 562:605–609.
3. John S, Chen H, Deng M, et al. A novel anti-LILRB4 CAR-T cell for the treatment of monocytic AML. Mol Ther. 2018; 26(10):2487-2495.
4. Cella M, Döhring C, Samaridis J, Dessing M, Brockhaus M, Lanzavecchia A, Colonna M. A novel inhibitory receptor (ILT3) expressedon monocytes, macrophages, and dendritic cells involvedin antigen processing. J Exp Med. 1997; 185(10):1743-51.
5. Kim-Schulze S, Scotto L, Vlad G, et al. Recombinant Ig-like transcript 3-Fc modulates T cell responses via induction of Th anergy and differentiationof CD8+ T suppressor cells. J Immunol. 2006;176(5):2790-8.
6. Paavola K, Roda JM, Lin VY, et al. The fibronectin-ILT3 interaction functions as a stromal checkpoint that suppresses myeloid cells. 2021, 9:1283-1297.